1104

Activity of the Multi-targeted Kinase Inhibitor, AT9283 on Imatinib-Resistant CML Models

Ruriko Tanaka[1], Matthew S Squires[2], Shinya Kimura[1], Asumi Yokota[1], Kirsty Mallett[2], Matthias Reule[2], Tomoko Smyth[2], Neil T Thompson[2], Rina Nagao[1], Takahiro Yamauchi[3], Eishi Ashihara[1], John F Lyons[2], Taira Maekawa[1]

[1]Department of Transfusion Medicine and Cell Therapy, Kyoto University Hospital, 54 Shogoin Kawahara-cho, Sakyo-ku, Kyoto, 606-8507, Japan. [2]Astex Therapeutics Ltd., 436 Cambridge Science Park, Milton Road, Cambridge, CB4 0QA, United Kingdom.
[3]Department of Hematology and Oncology, Faculty of Medical Sciences, University of Fukui, 23-3, Matsuokashimoaizuki, Eiheiji-cho, Yoshida-gun, Fukui 910-1193, Japan.

INTRODUCTION

- AT9283 is a potent inhibitor of JAK2, JAK3, mutant ABL kinase (T315I) and Aurora kinases A and B all of which have an IC50 < 5nM.
- CML is caused by a consistent genetic abnormality, termed the Philadelphia chromosome, that results from a reciprocal (9;22) translocation leading to the expression of the BCR-ABL fusion protein.
- Although the introduction of kinase inhibitors such as imatinib (Gleevec®) has revolutionised treatment of CML by targeting ABL activity, reactivation of the kinase via several different point mutations remains problematic.
- We describe here the characterisation of the anti-tumour effects of AT9283 in models of BCR-ABL dependent disease.
- AT9283 has potent anti-proliferative activity in a panel of Ba/F3 and human cell lines expressing the BCR-ABL fusion protein or its mutant forms including T315I.
- Treatment of several *in vivo* models of imatinib resistant CML resulted in significant growth inhibition and, in several cases, regression of the tumour. In some instances mice remained tumour free at 90 days following initial administration of compound, 72 days after the final administration.

COMPOUND PROFILE

A

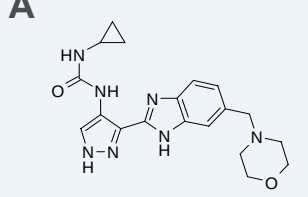

Chemical structure of AT9283

B

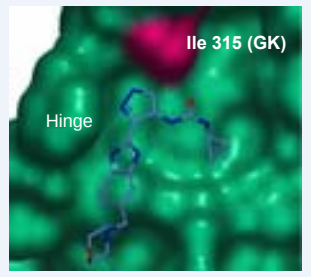
Ile 315 (GK)
Hinge

Model of AT9283 bound within the ATP binding site of T315I BCR-ABL

Howard et al., J Med Chem, in press

Figure 1: AT9283 Compound Structure

- Figure 1A shows the chemical structure of AT9283.
- Figure 1B is a model of AT9283 bound within the active site of T315I BCR-ABL. AT9283 does not make contact with threonine 315 in the same way that other kinase inhibitors in the class do.
- Similarly because the pocket behind the threonine 315 gatekeeper is not occupied by AT9283 there is no clash with this residue upon mutation to isoleucine.
- Hence AT9283 maintains activity vs the mutant forms of ABL. In fact, in *in vitro* assays at least, AT9283 appears to be more potent vs the mutant than the WT form (Table 1).
- Proliferation of a panel of Ba/F3 cell lines harbouring either wild type or various imatinib resistant mutant forms of the BCR-ABL kinase were inhibited with an IC_{50} of 10-21nM. (Table 2)
- Mutant forms of BCR-ABL that were inhibited include those highlighted in blue. These represent the most problematic resistant mutants in the clinic (Table 2).
- AT9283 is effective at inhibiting T315I-dependent Ba/F3 cell lines, the mutation resistant to all current ABL-kinase therapies. Similar data were obtained in human CML cell line models that are either sensitive, or have an acquired resistance to, imatinib therapy (Table 3).
- In certain cases, as indicated in Table 3, the dominant phenotype observed is that of polyploidy resulting from Aurora B inhibition. It is likely that we observe both Aurora and BCR-ABL inhibition in these cell lines and the predominant phenotype observed depends upon the specific genetic background of the cell in question.

AT9283 *IN VITRO* PROFILE

Protein Kinase	AT9283 IC_{50} (nM)
BCR-ABL (T315I)	4
BCR-ABL (WT)	110
JAK2	1.2
JAK3	1.1
Aurora A	52% @ 3nM
Aurora B	58% @ 3nM

Table 1: *in vitro* Kinase Inhibition Profile of AT9283

Ba/F3	AT9283 IC_{50} (nM)
WT p190	16
WT p210	13
Y253F	16
T315I	**11**
T315A	**10**
Q252H	21
M351T	18
M294V	18
H396P	21
G250E	12
F317V	**14**
F317L	**15**
E255K	**13**

Table 2: Activity of AT9283 in a Panel of BCR-ABL Ba/F3 Cell Lines

- Ba/F3 cells were engineered to stably express wild-type (WT) or mutant forms of BCR-ABL.
- Cells were exposed to AT9283 for 72h. Cell viability was determined using an Alamar Blue™ assay.

	Cell Line	BCR-ABL Status	Additional Characteristics	IC_{50} (nM)
Imatinib Sensitive	BV173	+		5.5
	KU812	+		26
	MYL	+		21
	KT-1	+		81
	KBM-5	+		84
	MEG-01	+		31
	K562	+		Polyploidy @ 30
Imatinib Resistant	HL60	-		Polyploidy @ 30
	KBM5-STIR	+ (T315I)		16
	K562/D1-9	+	PgP Overexpressor	>1000
	K562/Bcl-2	+	Bcl2 Overexpressor	50% @ 300
	K562/IMR	+	BCR-ABL overexpressor	64% @ 300
	K562/Bcl-xL	+	Bcl-xL Overexpressor	41% @ 300
	BV173/shBim	+	Bim Knockdown	12

Table 3: Activity of AT9283 in a Panel of Ph+ Human CML Cell Lines

- Human CML cell lines were exposed to AT9283 for 72h. Cell viability was determined using an Alamar Blue™ assay.

MECHANISM OF ACTION IN CML CELL LINES

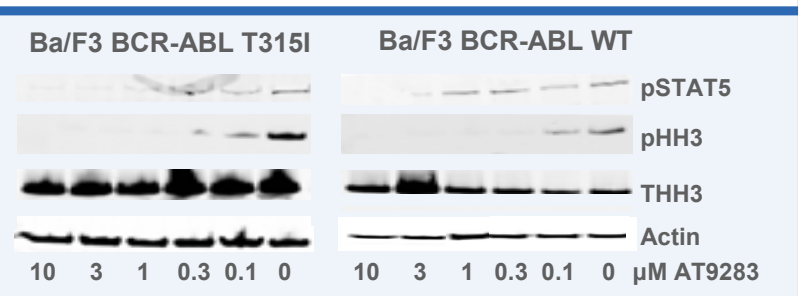
Ba/F3 BCR-ABL T315I
Ba/F3 BCR-ABL WT
pSTAT5
pHH3
THH3
Actin
10 3 1 0.3 0.1 0
10 3 1 0.3 0.1 0 µM AT9283

Figure 2: Mechanism of Action of AT9283 in Ba/F3 BCR-ABL Cells

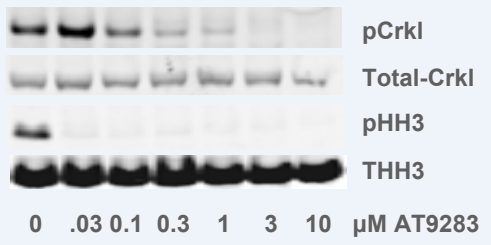
pCrkl
Total-Crkl
pHH3
THH3
0 .03 0.1 0.3 1 3 10 µM AT9283

Figure 3: Mechanism of Action of AT9283 in BV-173 CML Cells

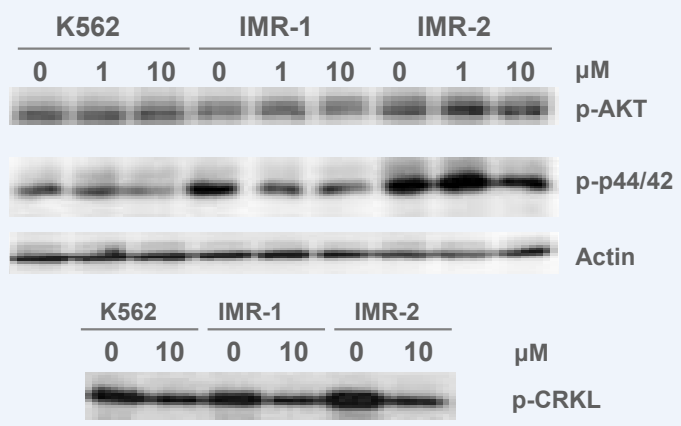
K562
IMR-1
IMR-2
0 1 10 0 1 10 0 1 10 µM
p-AKT
p-p44/42 MAPK
Actin
K562
IMR-1
IMR-2
0 10 0 10 0 10 µM
p-CRKL

Figure 4: Mechanism of Action of AT9283 in K562 CML Cells

- Ba/F3 BCR-ABL wild type or T315I mutant cells were treated with AT9283 for 4 hours (Figure 2). BV-173 CML cells that express wild-type BCR-ABL (Figure 3) and K562 WT BCR-ABL or two imatinib resistant forms (IMR-1 and IMR-2) were exposed to AT9283 for 24h. Lysates were harvested for western blotting.
- Following exposure to AT9283 downstream substrates of Aurora B (pHistone H3 (pHH3)) and BCR-ABL (pSTAT5, pCrkl) were inhibited (Figure 2, 3 and 4) in Ba/F3 cells and human CML cell lines including Ba/F3 cells harbouring T315I BCR-ABL.
- Inhibition of the Aurora substrate pHH3 was observed at roughly 10 fold lower concentrations than the substrates of WT BCR-ABL consistent with the *in vitro* activity of AT9283 vs these targets (Table 1).
- These data suggest that both the Aurora and BCR-ABL inhibitory effects of AT9283 manifest themselves in CML cell lines and that either or both activities could result in the efficacy of the compound in these CML models.

IN VIVO ACTIVITY IN CML MODELS

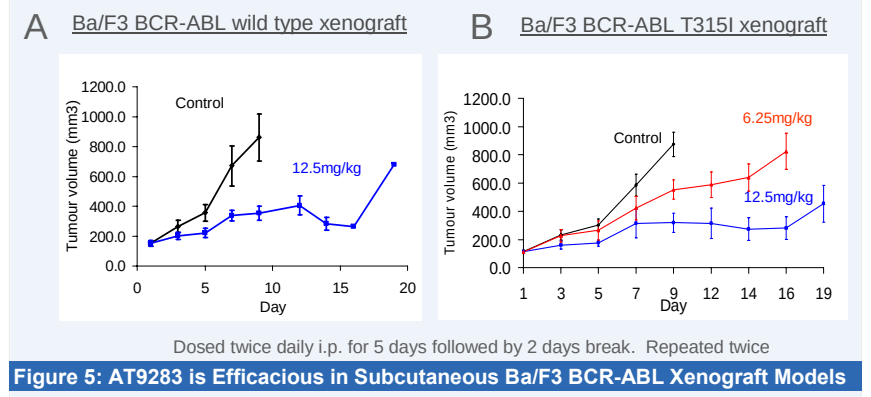
A
Ba/F3 BCR-ABL wild type xenograft
B
Ba/F3 BCR-ABL T315I xenograft
Tumour volume (mm3)
Control
12.5mg/kg
6.25mg/kg
Day
Dosed twice daily i.p. for 5 days followed by 2 days break. Repeated twice

Figure 5: AT9283 is Efficacious in Subcutaneous Ba/F3 BCR-ABL Xenograft Models

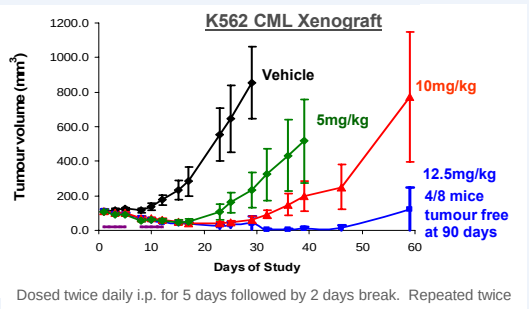
K562 CML Xenograft
Tumour volume (mm3)
Vehicle
5mg/kg
10mg/kg
12.5mg/kg
4/8 mice
tumour free
at 90 days
Days of Study
Dosed twice daily i.p. for 5 days followed by 2 days break. Repeated twice

Figure 6: AT9283 is Efficacious in Subcutaneous K562 Human CML Xenograft Model

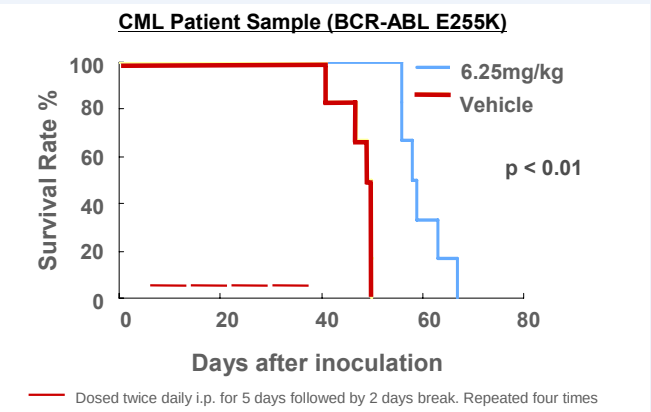
CML Patient Sample (BCR-ABL E255K)
Survival Rate %
6.25mg/kg
Vehicle
p < 0.01
Days after inoculation
Dosed twice daily i.p. for 5 days followed by 2 days break. Repeated four times

Figure 7: AT9283 is Efficacious in a Primary Cell BCR-ABL (E255K) Xenograft Model

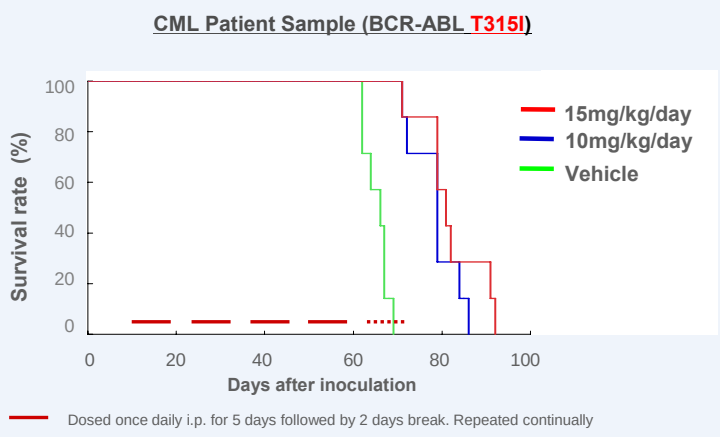
CML Patient Sample (BCR-ABL T315I)
Survival rate (%)
15mg/kg/day
10mg/kg/day
Vehicle
Days after inoculation
Dosed once daily i.p. for 5 days followed by 2 days break. Repeated continually

Figure 8: AT9283 is Efficacious in a Primary Cell BCR-ABL (T315I) Xenograft Model

IN VIVO ACTIVITY IN CML MODELS

- In each of the studies shown tumour-bearing nude mice were dosed via the i.p route once or twice daily for 5 days in 7, repeated for the indicated number of cycles.
- The volume of the s.c. tumour was calculated as an ellipsoid volume every 2 days (Fig 5 and 6).
- Figures 5A and 5B show that AT9283 inhibits tumour growth in subcutaneous xenograft models with Ba/F3 cells transfected with either wild type BCR-ABL kinase or the T315I mutant form.
- Figure 6 shows that AT9283 induced prolonged inhibition of tumour growth and regression in a human CML xenograft model, K562 cells.
- At 12.5mg/kg regressions were observed and 50% of the mice in this dose group remained tumour free out to 90 days, 78 days after administration of the final dose of AT9283.
- In the case of the primary CML models male NOD/SCID mice were sub-lethally irradiated and inoculated intravenously with leukaemic cells harbouring BCR-ABL E255K and T315I. (Figures 7 and 8)
- Kaplan Meier survival curves show that treatment with 6.25mg/kg AT9283 twice daily resulted in a significant survival advantage (p=0.008) over vehicle-treated animals of 17 days in the E255K model. Similarly, in the T315I model 10mg/kg/day or 15mg/kg/day AT9283 resulted in a marked survival advantage (p=0.002).

CONCLUSIONS

- AT9283 is a multi-targeted kinase inhibitor with activity against Aurora A and B and BCR-ABL including many of the identified mutants forms.
- AT9283 inhibits survival of engineered cell lines expressing BCR-ABL or its mutant forms as well as human leukaemia cells harbouring the same mutations.
- These antiproliferative effects can be attributed to inhibition of BCR-ABL in addition to activity vs other targets in CML cell lines.
- AT9283 exhibited potent inhibitory effects in animal models with either Ba/F3 cells, human leukaemic cell lines or primary human CML samples.
- These data support further clinical investigation of AT9283 in patients with treatment resistant CML.

Disclosure Statements

Research Support/P.I.	No relevant conflicts of interest to declare
Employee	M.S., K.M., T.M., N.T., J.L. (Astex Therapeutics Ltd.)
Consultant	No relevant conflicts of interest to declare
Major Stockholder	No relevant conflicts of interest to declare
Speakers' Bureau	No relevant conflicts of interest to declare
Honoraria	No relevant conflicts of interest to declare
Scientific Advisory Board	No relevant conflicts of interest to declare

Presented at the American Society for Hematology 50th Annual Meeting and Exposition, December 6th-9th, 2008, San Francisco CA.

This poster is available to download at www.astex-therapeutics.com

© 2008 Astex Therapeutics Limited. All rights reserved